5-Year Surveillance (2003-2007) of Anti-pneumococcal Activity of Oral Agents Recommended for the Empirical Treatment of Community Acquired Pneumonia (CAP) in Adults

Presentation No. C2-204

Y. C. Yee[1], A. T. Evangelista[1], M. Obot-Tucker[2], N. P. Brown[2], C. M. Pillar[2], C. Thornsberry [2], D. F. Sahm [2], and S. Graff[2]
[1]Ortho-McNeil, Inc., Raritan, NJ, USA, [2]Eurofins Medinet Inc., Anti-Infective Services, Herndon, VA USA

Contact Information:
Daniel Sahm, Ph.D.
Eurofins Medinet, Inc., Anti-Infective Services
Herndon, VA, USA
dan.sahm@eurofinsmedinet.com

ABSTRACT

BACKGROUND: Based on various criteria, recent guidelines for the empirical treatment of CAP encompass the use of one or more of the following; macrolides, doxycycline, β-lactams, and respiratory fluoroquinolones. This study analyzed 5 years of data from an ongoing surveillance initiative to identify trends in activity between *Streptococcus pneumoniae* (SP) and recommended oral agents.

METHODS: SP (n = 21,400) respiratory isolates from TRUST surveillance 2003-2007 were centrally tested by broth microdilution (CLSI M7-A6; M100-S16). Results for amoxicillin-clavulanate (AMX), azithromycin (AZM), cefuroxime-axetil (CXM), levofloxacin (LVX), moxifloxacin (MXF), tetracycline (TET), and penicillin (PEN) were included.

RESULTS: From 2003-2007, susceptibility of SP to PEN was relatively stable (64.0-66.3%) with a constant MIC_{90} of 2 µg/ml. CXM resistance fluctuated between 20.7% (2003) and 18.5% (2007) with MIC_{90}s ≥ 4 µg/ml. AMX resistance increased from 4.1% (2003) to 9.1% (2007) and the MIC_{90} increased from 2 to 4 µg/ml. AZM resistance increased from 27.5% (2003) to 32.3 % (2007); from 2004 to 2005 the MIC_{90} increased from 16 to >128 µg/ml and remained high into 2007. TET resistance increased from 13.5% (2004) to 18.8% (2007) with a constant MIC_{90} of 32 µg/ml. Except for 2004, the % susceptibility of MXF and LVX was >99%; MXF and LVX MIC_{90}s remained constant at 0.25 µg/ml and 1 µg/ml, respectively.

CONCLUSION: Trends indicate that SP penicillin resistance has slightly decreased, while resistance rates for AMX increased from 4.1% in 2003 to 9.1% in 2007. Increased resistance to AZM and TET along with high MIC_{90}s indicate that continued use of macrolides and tetracyclines could be problematic. The activities of respiratory fluoroquinolones remained high, and no trends that would indicate changes in activity were detected.

BACKGROUND

In 2007, the American Thoracic Society (ATS) and the Infectious Diseases Society of America (IDSA) issued consensus guidelines for the management and treatment of community-acquired pneumonia (CAP). Empiric treatment is directed by patient location (outpatient vs inpatient), and likelihood of the involvement of drug resistant *S. pneumoniae* based on risk factors and local resistance data. The use of respiratory fluoroquinolones (FQs), levofloxacin (LVX) and moxifloxacin (MXF), has been recommended for CAP patients with comorbidities, with recent antibiotic exposure, or in regions where high level resistance to macrolides (MIC ≥ 16 µg/ml) among *S. pneumoniae* is documented to occur at high frequency (>25%). Previous studies of respiratory FQs have shown high activity against *S. pneumoniae*, and this study reviews data from the last 5 years (2003-2007) comparing these agents with other recommended oral agents. In addition, the pharmacodynamic (PD) activity of the respiratory FQs is examined, since AUC/ MIC_{90} ratios above 30 for *S. pneumoniae* are considered to be good predictors of successful clinical outcomes, microbiologic eradication, and reduced potential for the emergence of resistance (Zhanel et al. *JAC* 2001;47:435). For the purpose of comparison, ciprofloxacin, an agent which is not recommended for treatment of CAP by the ATS/IDSA, was added to the PD analysis.

Table 1. Susceptibility of *S. pneumoniae* (TRUST 7-11 [2003-2007])

	TRUST 7 (2003) n=4452			TRUST 8 (2004) n=4338			TRUST 9 (2005) n= 4958			TRUST 10 (2006) n= 3932			TRUST 11 (2007) n=3720		
Antimicrobial agent	%S	%I	%R	%S	%I	%R	%S	%I	%R	%S	%I	%R	%S	%I	%R
Amoxicillin/ Clavulanate	93.4	2.5	4.1	91.8	3.0	5.2	91.8	1.9	6.3	91.2	2.2	6.6	88.2	2.7	9.1
Azithromycin	72.2	0.3	27.5	74.1	0.8	25.1	70.7	0.5	28.8	67.6	0.5	31.9	67.4	0.3	32.3
Cefuroxime/ Axetil	76.1	3.2	20.7	77.5	2.1	20.4	80.0	2.6	17.5	78.1	4.1	17.8	78.0	3.4	18.5
Levofloxacin	99.1	0.0	0.9	98.7	0.2	1.1	99.2	0.0	0.8	99.4	0.1	0.6	99.1	0.2	0.7
Moxifloxacin	99.2	0.4	0.4	98.9	0.4	0.6	99.3	0.3	0.5	99.4	0.1	0.4	99.3	0.5	0.2
Penicillin	66.3	16.4	17.3	65.2	16.0	18.8	65.1	19.3	15.6	62.9	22.6	14.5	64.0	22.7	13.3
Trimeth/ Sulfa	70.3	5.8	23.9	72.1	6.6	21.3	73.2	6.5	20.3	71.0	7.5	21.5	72.2	7.7	20.1
Tetracycline	NT	NT	NT	86.2	0.2	13.5	84.6	0.3	15.2	82.9	0.2	16.9	80.8	0.5	18.8

NT: Not tested

Table 2. Activity of oral agents against *S. pneumoniae* (TRUST 7-11 [2003-2007])

	TRUST 7 (2003) n=4,452 MIC (µg/ml)			TRUST 8(2004) n= 4,338 MIC (µg/ml)			TRUST 9(2005) n=4,958 MIC (µg/ml)			TRUST 10(2006) n= 3,932 MIC (µg/ml)			TRUST 11(2007) n= 3,720 MIC (µg/ml)		
Antimicrobial agent	Mode	MIC_{50}	MIC_{90}	Mode	MIC_{50}	MIC_{90}	Mode	MIC_{50}	MIC_{90}	Mode	MIC_{50}	MIC_{90}	Mode	MIC_{50}	MIC_{90}
Amoxicillin/ Clavulanate	≤0.03	≤0.03	2	0.03	0.03	2	0.03	0.03	2	0.03	0.03	2	0.03	0.03	4
Azithromycin	≤0.06	≤0.06	16	0.06	0.06	16	0.12	0.12	>128	0.06	0.12	>128	0.06	0.12	>128
Cefuroxime/ Axetil	≤0.25	≤0.25	>4	0.03	0.03	4	0.03	0.06	4	0.03	0.06	4	0.03	0.06	8
Levofloxacin	1	1	1	1	1	1	1	1	1	1	1	1	1	1	1
Moxifloxacin	0.12	0.12	0.25	0.12	0.12	0.25	0.12	0.12	0.25	0.12	0.12	0.25	0.12	0.12	0.25
Penicillin	≤0.03	≤0.03	2	≤0.015	≤0.015	2	≤0.015	≤0.015	2	≤0.015	0.03	2	≤0.015	0.03	2
Trimeth/ Sulfa	0.12	0.25	>4	0.25	0.25	8	0.25	0.25	4	0.25	0.25	4	0.12	0.25	4
Tetracycline	NT	NT	NT	0.25	0.5	32	0.25		>8	0.25	0.25	32	0.25	0.25	32

NT: Not tested

Figure 1. Changes in percent resistant isolates over time of oral CAP agents against *S. pneumoniae* 2003-2007

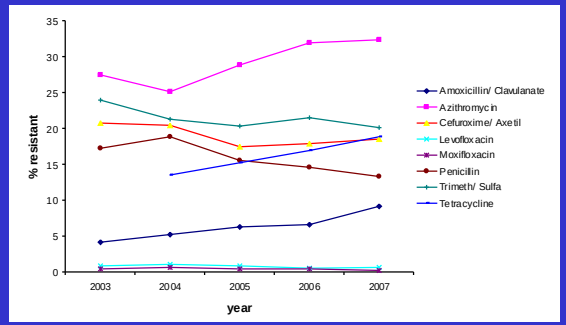
35
30
25
20
15
10
5
0
% resistant
2003
2004
2005
2006
2007
year
Amoxicillin/ Clavulanate
Azithromycin
Cefuroxime/ Axetil
Levofloxacin
Moxifloxacin
Penicillin
Trimeth/ Sulfa
Tetracycline

Table 3. Pharmacodynamic Activity of Fluoroquinolones against *S. pneumoniae*

Antimicrobial (single dose, PO)	MIC_{90} [a]	AUC_{0-24h} total/free (mg•h/mL) [b]	AUC_{0-24h} free/ MIC_{90}
Levo (500 mg qd)	1	48.0/33.1	33
Levo (750 mg qd) [c]	1	101.0/70.7	71
Moxi (400 mg qd)	0.25	38.1/17.9	72
Cipro (500 mg q12h)	2	20.2/14.1	7

[a]TRUST 11 (2006-2007), Data on file, Ortho-McNeil, Inc.
[b]Zhanel et al. *Drugs*. 2002;62:13-59. Area under the serum concentration-time curve following single PO dose in normal volunteers. Free AUC based on 31% protein binding for levofloxacin, 30% for ciprofloxacin, and 47% for moxifloxacin.
[c]LEVAQUIN® Prescribing Information.

METHODS

During 2003-2007, a total of 21,400 respiratory isolates of *S. pneumoniae* were collected from laboratories across the 9 US Bureau of Census regions for the TRUST surveillance initiative. These isolates were identified and tested by a central lab, Eurofins Medinet, Inc., using standard reference methodologies. Antimicrobial agents of clinical interest in the treatment of respiratory infections were tested including: amoxicillin/clavulanate, azithromycin, cefuroxime/axetil, levofloxacin, moxifloxacin, penicillin, trimethoprim/sulfamethoxazole, and tetracycline. These agents were tested against *S. pneumoniae* isolates by broth microdilution according to CLSI methodology, M7-A6. MIC results were interpreted as resistant, intermediate, or susceptible according to CLSI document M100-S17.

RESULTS

•Resistance (R) rates to 4 common oral agents increased during the 5-year study period. From 2003 to 2007, azithromycin-R increased from 27.5% to 32.3% and amoxicillin/clavulanate-R increased from 4.1% to 9.1% **(Table 1, Figure 1)**. In addition, tetracycline-R increased from 13.5% to 18.8% (2004-2007) and cefuroxime-R increased from 17.5% to 18.5% (2005-2007) **(Table 1, Figure 1)**.

•There was a large increase in the MIC_{90} of azithromycin from 2004 (16 µg/ml) to 2005 (>128 µg/ml) **(Table 2)**, and higher MIC_{90}s for azithromycin were then detected in 2006 and 2007 (>128 µg/ml) **(Table 2)**. From 2006 to 2007, the MIC_{90} of amoxicillin/clavulanate increased from 2 to 4 µg/mL and for cefuroxime from 4 to 8 µg/mL **(Table 2)**.

•There was a slight decrease in the amount of penicillin resistant isolates (MIC ≥ 2 µg/mL) over time (17.3% in 2003 to 13.3% in 2007) **(Table 1, Figure 1)**.

•The MIC_{90} of the tested respiratory fluoroquinolones (levofloxacin and moxifloxacin) remained constant (1 µg/ml for levofloxacin and 0.25 µg/ml for moxifloxacin) from 2003 to 2007 **(Table 2)**. Greater than 99% of isolates were susceptible to both levofloxacin and moxifloxacin for all years tested with the exception of 2004 (levofloxacin %S = 98.7%, moxifloxacin %S = 98.9%) **(Table 1)**. Ciprofloxacin, for the purpose of comparison, differed from the respiratory fluoroquinolones with decreasing susceptibility rates of 86.7% in 2005 to 82.0% in 2007 **(Data on file)**. Since ciprofloxacin does not have CLSI breakpoints for *S. pneumoniae*, FDA breakpoints were used for the surveillance study.

•The AUC_{0-24h}/MIC_{90} ratios for the respiratory fluoroquinolones (levofloxacin and moxifloxacin) exceeded 30 (the target PD ratio that correlates with clinical cure, microbiological eradication, and prevention of the emergence of resistance [Zhanel et al, *JAC* 2001;47:435]) **(Table 3)**. Nearly identical AUC_{0-24h}/MIC_{90} were obtained for levofloxacin 750mg qd (71) and moxifloxacin 400mg qd (72) **(Table 3)**. The ratio (7) of ciprofloxacin (500mg q12h) was < ¼ of the target PD **(Table 3)**.

CONCLUSION

•Resistance rates to 4 common oral agents increased during the 5-year study period: azithromycin (32.3%), amoxicillin/clavulanate (9.1%), cefuroxime (18.5%), and tetracycline (18.8%). These increasing resistance trends for these agents suggest that caution should be used when treating respiratory infections caused by *S. pneumoniae*.

•The respiratory fluoroquinolones (levofloxacin and moxifloxacin) were the most active agents *in vitro* against *S. pneumoniae* from 2003-2007, as reflected by low MIC_{90}s and high percent susceptibilities.

•Of the fluoroquinolones, levofloxacin and moxifloxacin provided pharmacodynamic activity above the target PD ratio of 30 (AUC_{0-24h} free/MIC_{90}) for *S. pneumoniae*. Ciprofloxacin is not recommended for the treatment of CAP by ATS/IDSA, and the PD ratio of 7 (higher potential for emergence of resistance, and lower potential for successful clinical outcome and microbiologic eradication) supports this guideline.

ACKNOWLEDGEMENTS

This study was supported by Ortho-McNeil, Inc., Raritan, NJ

47th Interscience Conference on Antimicrobial Agents and Chemotherapy (ICAAC)
Chicago, IL
September 17-20, 2006